Identification and Characterization of HCV Replicon Variants with Reduced Susceptibility to HCV-796

Anita Y. M. Howe,[1] Huiming Cheng,[1] Stephan Johann,[1] Stanley Mullen,[1] Srinivas K. Chunduru,[2] Dorothy C. Young,[2] Joel Bard,[1] Rajiv Chopra,[1] and John O'Connell[1]

[1]Wyeth Research, Collegeville, PA; [2]ViroPharma Incorporated, Exton, PA

ABSTRACT

HCV-796 has been shown to selectively inhibit hepatitis C virus (HCV) NS5B RNA dependent RNA polymerase with an IC_{50} of 40 nM in a biochemical assay. In hepatoma cells containing a subgenomic genotype 1b HCV replicon, HCV-796 reduced HCV RNA levels by 3-4 $\log_{10}$ HCV copies/µg total RNA (EC_{50}=9 nM). Cells bearing replicon variants with reduced susceptibility to HCV-796 were generated in the presence of HCV-796 followed by G418 selection. The variant cells displayed 23- to 6812-fold resistance to HCV-796. Sequence analysis of the NS5B gene derived from the replicon variants revealed several amino acid changes within 5 Å of the drug-binding pocket. Specifically, mutations at leucine 314, cysteine 316, isoleucine 363, serine 365, and methionine 414 of NS5B, which have been shown to directly interact with HCV-796, were observed. The impact of the amino acid substitutions on viral fitness and drug susceptibility was examined in recombinant replicons and NS5B enzymes molecularly engineered with the single amino acid mutations. The replicon variants were 10- to 100-fold less efficient in forming colonies in human hepatoma cells compared with the wild-type replicon; the S365 variant failed to establish a stable cell line. Other variants (L314F, I363V, and M414V) also had 4- to 9-fold lower steady state HCV RNA levels. While different levels of resistance to HCV-796 were observed in the replicon and enzyme variants, these variants retained their susceptibility to pegylated interferon (PegIFN), ribavirin, and other HCV specific inhibitors.

Selection of Replicon Variants

FIGURE 1.
Generation of HCV-796 Associated Replicon Variants

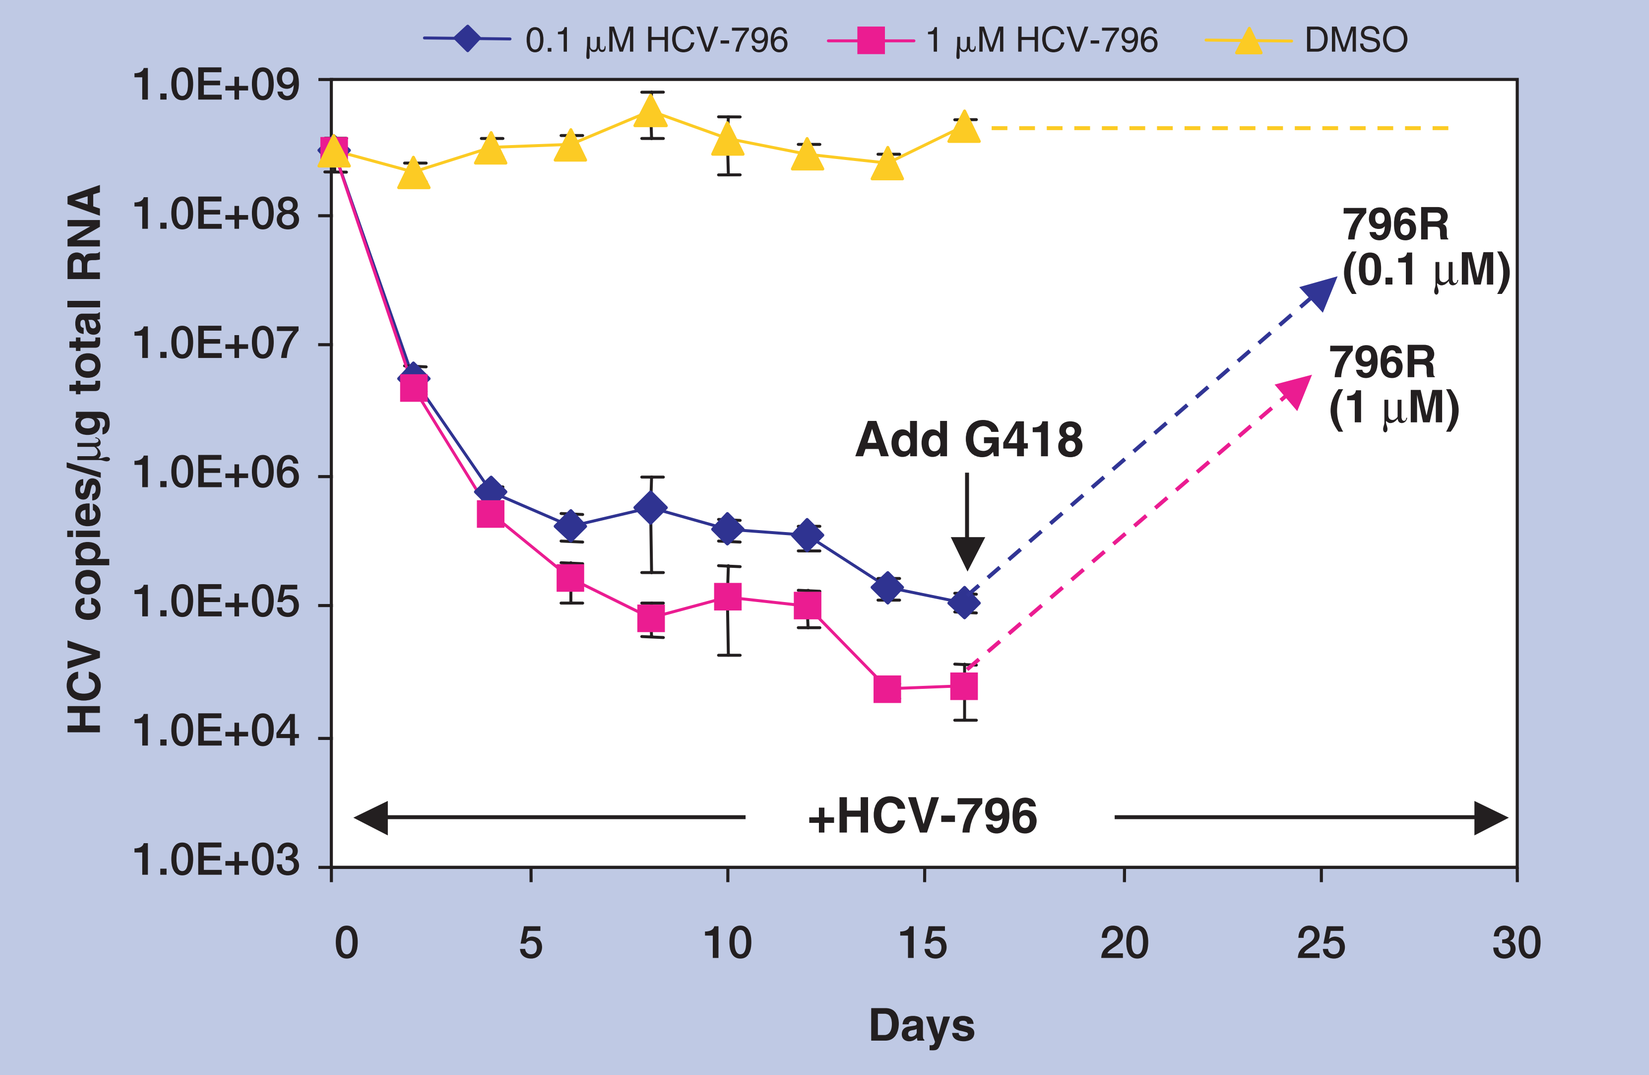

0.1 µM HCV-796
1 µM HCV-796
DMSO
1.0E+09
1.0E+08
1.0E+07
1.0E+06
1.0E+05
1.0E+04
1.0E+03
HCV copies/µg total RNA
796R (0.1 µM)
796R (1 µM)
Add G418
+HCV-796
0
5
10
15
20
25
30
Days

Replicon cells were treated with HCV-796 at concentrations equivalent to 10X (0.1 µM) and 100X (0.1 µM) EC_{50} in the absence of G416 in the first 16 days. Resistant replicons were selected with an addition of G418 and HCV-796. Broken arrows indicate the enrichment of replicons. A third pool of resistant cells was generated by further treating the cells with 10 µM HCV-796 [796R (10 µM)].

Drug Susceptibility of Replicon Cells Selected by HCV-796

FIGURE 2.
Antiviral Activities of HCV-796 in the Pools of Replicon Cells After Selection with HCV-796

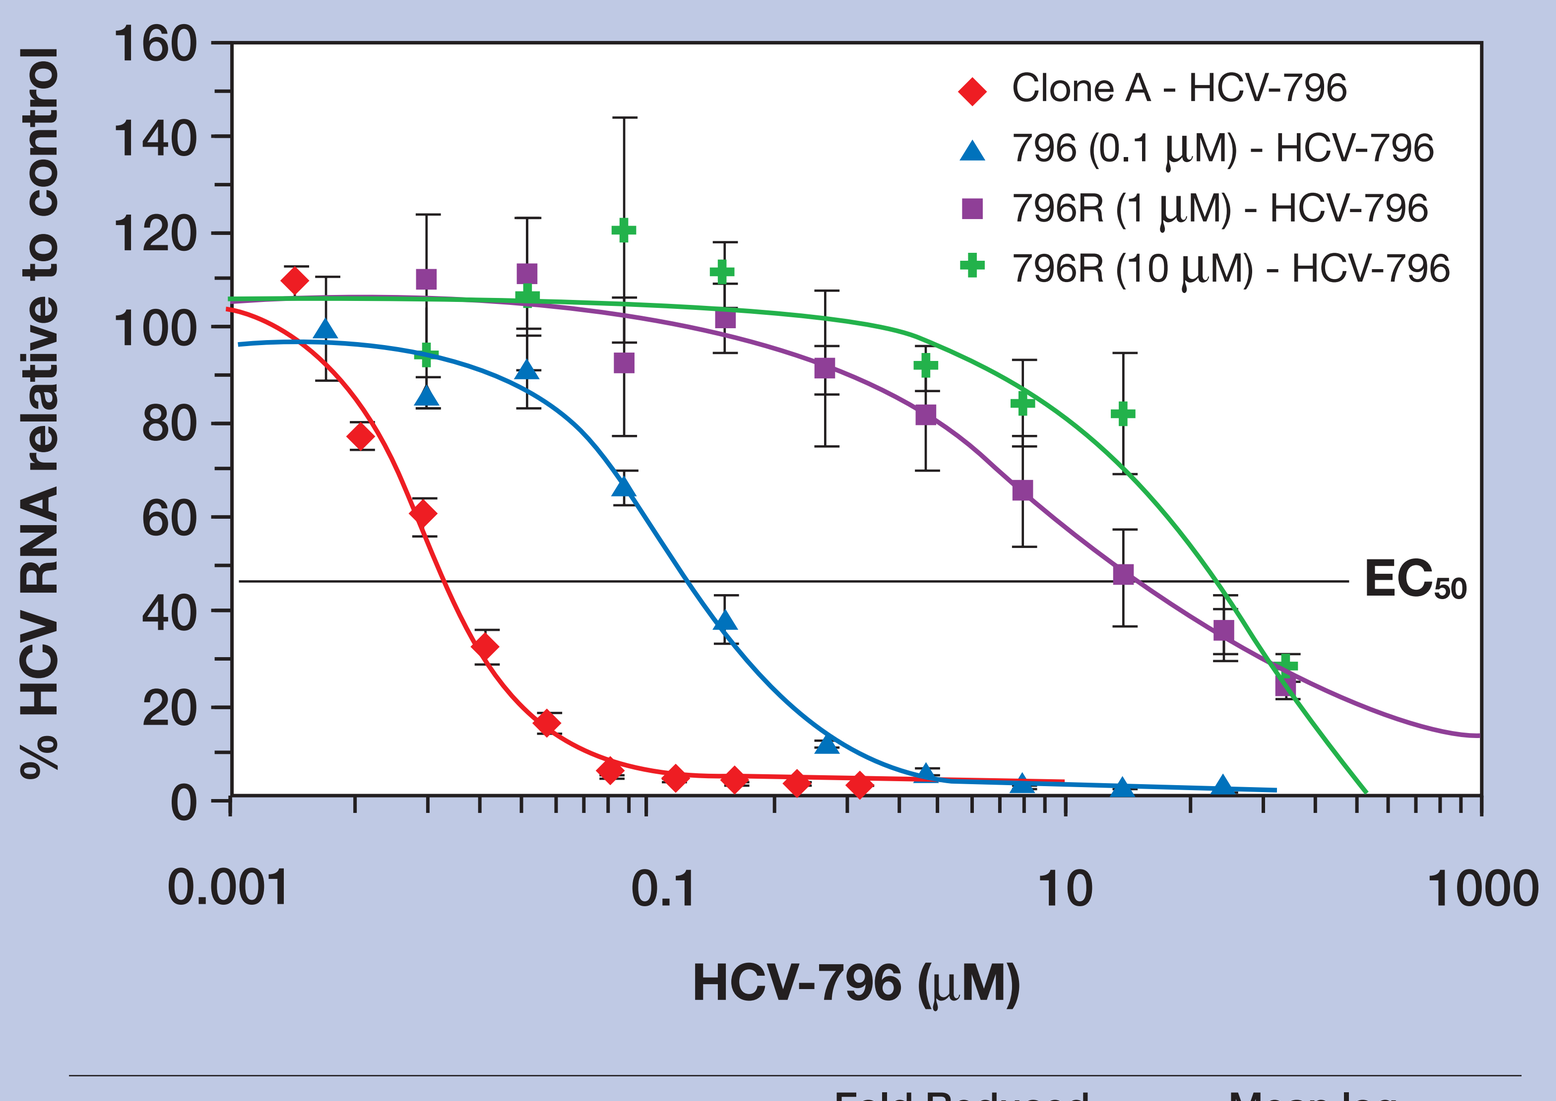

Clone A - HCV-796
796 (0.1 µM) - HCV-796
796R (1 µM) - HCV-796
796R (10 µM) - HCV-796
% HCV RNA relative to control
160
140
120
100
80
60
40
20
0
EC50
0.001
0.1
10
1000
HCV-796 (µM)

Cells[a]	EC_{50} (µM) ± SD[b]	Fold Reduced Susceptibility	Mean $\log_{10}$ Viral Reduction[c]
Clone A	0.013 ± 0.013 (n=8)	-	2.1 ± 0.4 (0.7 µM)
796R (0.1 µM)	0.3 ± 0.2 (n=4)	23	1.4 ± 0.3 (56 µM)
796R (1 µM)	8.0 ± 5.2 (n=12)	618	0.7 ± 0.2 (56 µM)
796R (10 µM)	>88.0 (n=4)	> 6812	0.5 ± 0.3 (56 µM)

a: 796R represents cells that are less susceptible to HCV-796. Concentrations of HCV-796 used for the selection are indicated in parentheses.
b: EC_{50} values were determined using the MDL LSW data analysis™. Inhibitory activity is expressed as mean EC_{50} ± standard deviation. n indicates number of determinations.
c: Viral load reduction was determined at the indicated compound concentrations in parentheses in a 3-day assay. Data represent the mean log reduction of viral RNA ± standard deviation. Results represent at least 3 independent determinations.

Mapping of Replicon Variants

Table 1a. Amino acid changes in NS5B derived from control cells

amino acid	5	53	57	79	114	116	117	122	125	140	182	231	253	322	345	353	369	407	412	424	432	443	465	471	478	531	533	556
pBB7 (genebank seq)	T	T	L	K	K	V	N	V	D	A	L	N	I	V	R	P	N	S	I	I	I	L	R	A	S	R	K	S
p7-1						I									G			P			V						E	
p7-2	A								N		P													T		G		
p7-3		A			R		S												T									
p7-4				R								D					S			T								
p7-5				R								D					S			T								
p7-6																				V	V				P			
p7-7								A					T	A														
p7-8										V													Q					
p7-9																L												G
p7-10			P																									
p7-11																						P						

pBB7 represents the input NS5B sequence in control cells

Table 1b. Amino acid changes in NS5B derived from HCV-796 associated replicon variant cells (796R)

Clones isolated from	amino acid	4	19	85	97	147	162	181	197	201	286	316	329	355	363	365	368	390	414	440	442	445	514	534	581
	pBB7	Y	T	V	A	V	F	T	P	V	T	C	T	K	I	S	S	T	M	E	A	C	Q	L	V
0.1 µM HCV-796	p1-4													R								F		F	
	p1-10													R		A						F		F	
	p1-26													R					T			F		V	
	p1-27													R								F		V	
	p1-25													R								F		F	
	p3-8																		I			F			
	p3-22																		I			F			
	p3-23																		I			F			
	p3-2														V							F			
	p3-11														V							F			
	p3-5									A												F			
	p3-14									A												F			
	p1-5																					F			
	p1-28																					F			
	p3-1																					F			
	p1-1																					F			
	p1-2																					F			
	p1-3																					F			
	p3-4																					F			
	p3-28																					F			
1 µM HCV-796	p4-2		P	L			Y					F													
	p4-3		P	L			Y					F													
	p4-4		P	L			Y					F													
	p4-5			L			Y					F													
	p4-7		P	L			Y					F													
	p4-8			L			Y					F													
	p4-10			L			Y					F													
	PCR4			L			Y					F													
	PCR4M			L			Y					F													
	p4-9			L			Y									L									
	p6-10															L									
	PCR6								A/P							L/S						C/F			A/V
	p9-8															A						F			
	p9-9															T						F			
	p6-1								A													F			A
	p6-2								A													F			A
	p6-3								A													F			A
	p6-11								A													F			A
	p6-26								A													F			A
	p6-8								A	A												F			A
	p6-12								A													F			
	p9-2									A												F			
	p9-13									A		S										F			
	p9-1		P									S										F			
	p9-3											S										F			
	p9-4		P									S										F			
	p9-7		P									S										F			
	PCR9											S/C										F			
	p9-5													R	V							F			
	p9-6													R	V							F			
	p4-14															L									
	p6-4															L									
	p6-7															L									
	p9-14																					F			
10 µM HCV-796	p41-1											Y							I		T				
	p41-2											Y							I		T				
	p41-3											Y							I						
	p41-5											Y							I						
	p41-15											Y							I						
	PCR41											C/Y							I						
	p41-7											Y							I	G					
	p41-17																		I						
	p49-3											Y		R								F			
	p41-4											Y										F			
	p41-6		A									Y										F			
	p44-14		P	L			Y					F													
	p44-15		P	L			Y					F													
	p44-20		P	L			Y					F													
	p44-32		P	L			Y					F													
	p44-16		P	L	L		Y					F											R		
	p44-23			L			Y					F													
	p44-13			L			Y					F													
	PCR44			L			Y/F					C/F				L/S									
	p46-15	H				I		I					I			L	F	I							
	p46-16	H				I		I					I			L	F	I							
	p49-5												I	R		L						F			
	PCR46													R		L		I							
	p46-7							I								L		I							
	p46-14							I								L		I							
	p46-8															L		I	T						
	p46-12										A					L		I		G					
	p46-9										M					L		I							
	p46-11															L		I							
	p46-13															L		I							
	p44-18															L						F	R		
	p41-13				V									R		T						F			
	p49-1															T						F			
	p49-4															T						F			
	p49-2		A											R		A						F			
	PCR49													R		L/S						F			
	p44-12															L									
	p44-22															L									
	p46-10															L									
No. of clones out of 93 clones		2	14	18	2	2	18	4	8	5	3	33	3	13	4	29	2	10	13	2	2	50	2	5	7
Frequency of amino acid substitution		2.2	15.1	19.4	2.2	2.2	19.4	4.3	8.6	5.4	3.2	35.5	3.2	14.0	4.3	31.2	2.2	10.8	14.0	2.2	2.2	53.8	2.2	5.4	7.5

Note: L314F was identified in a separate experiment

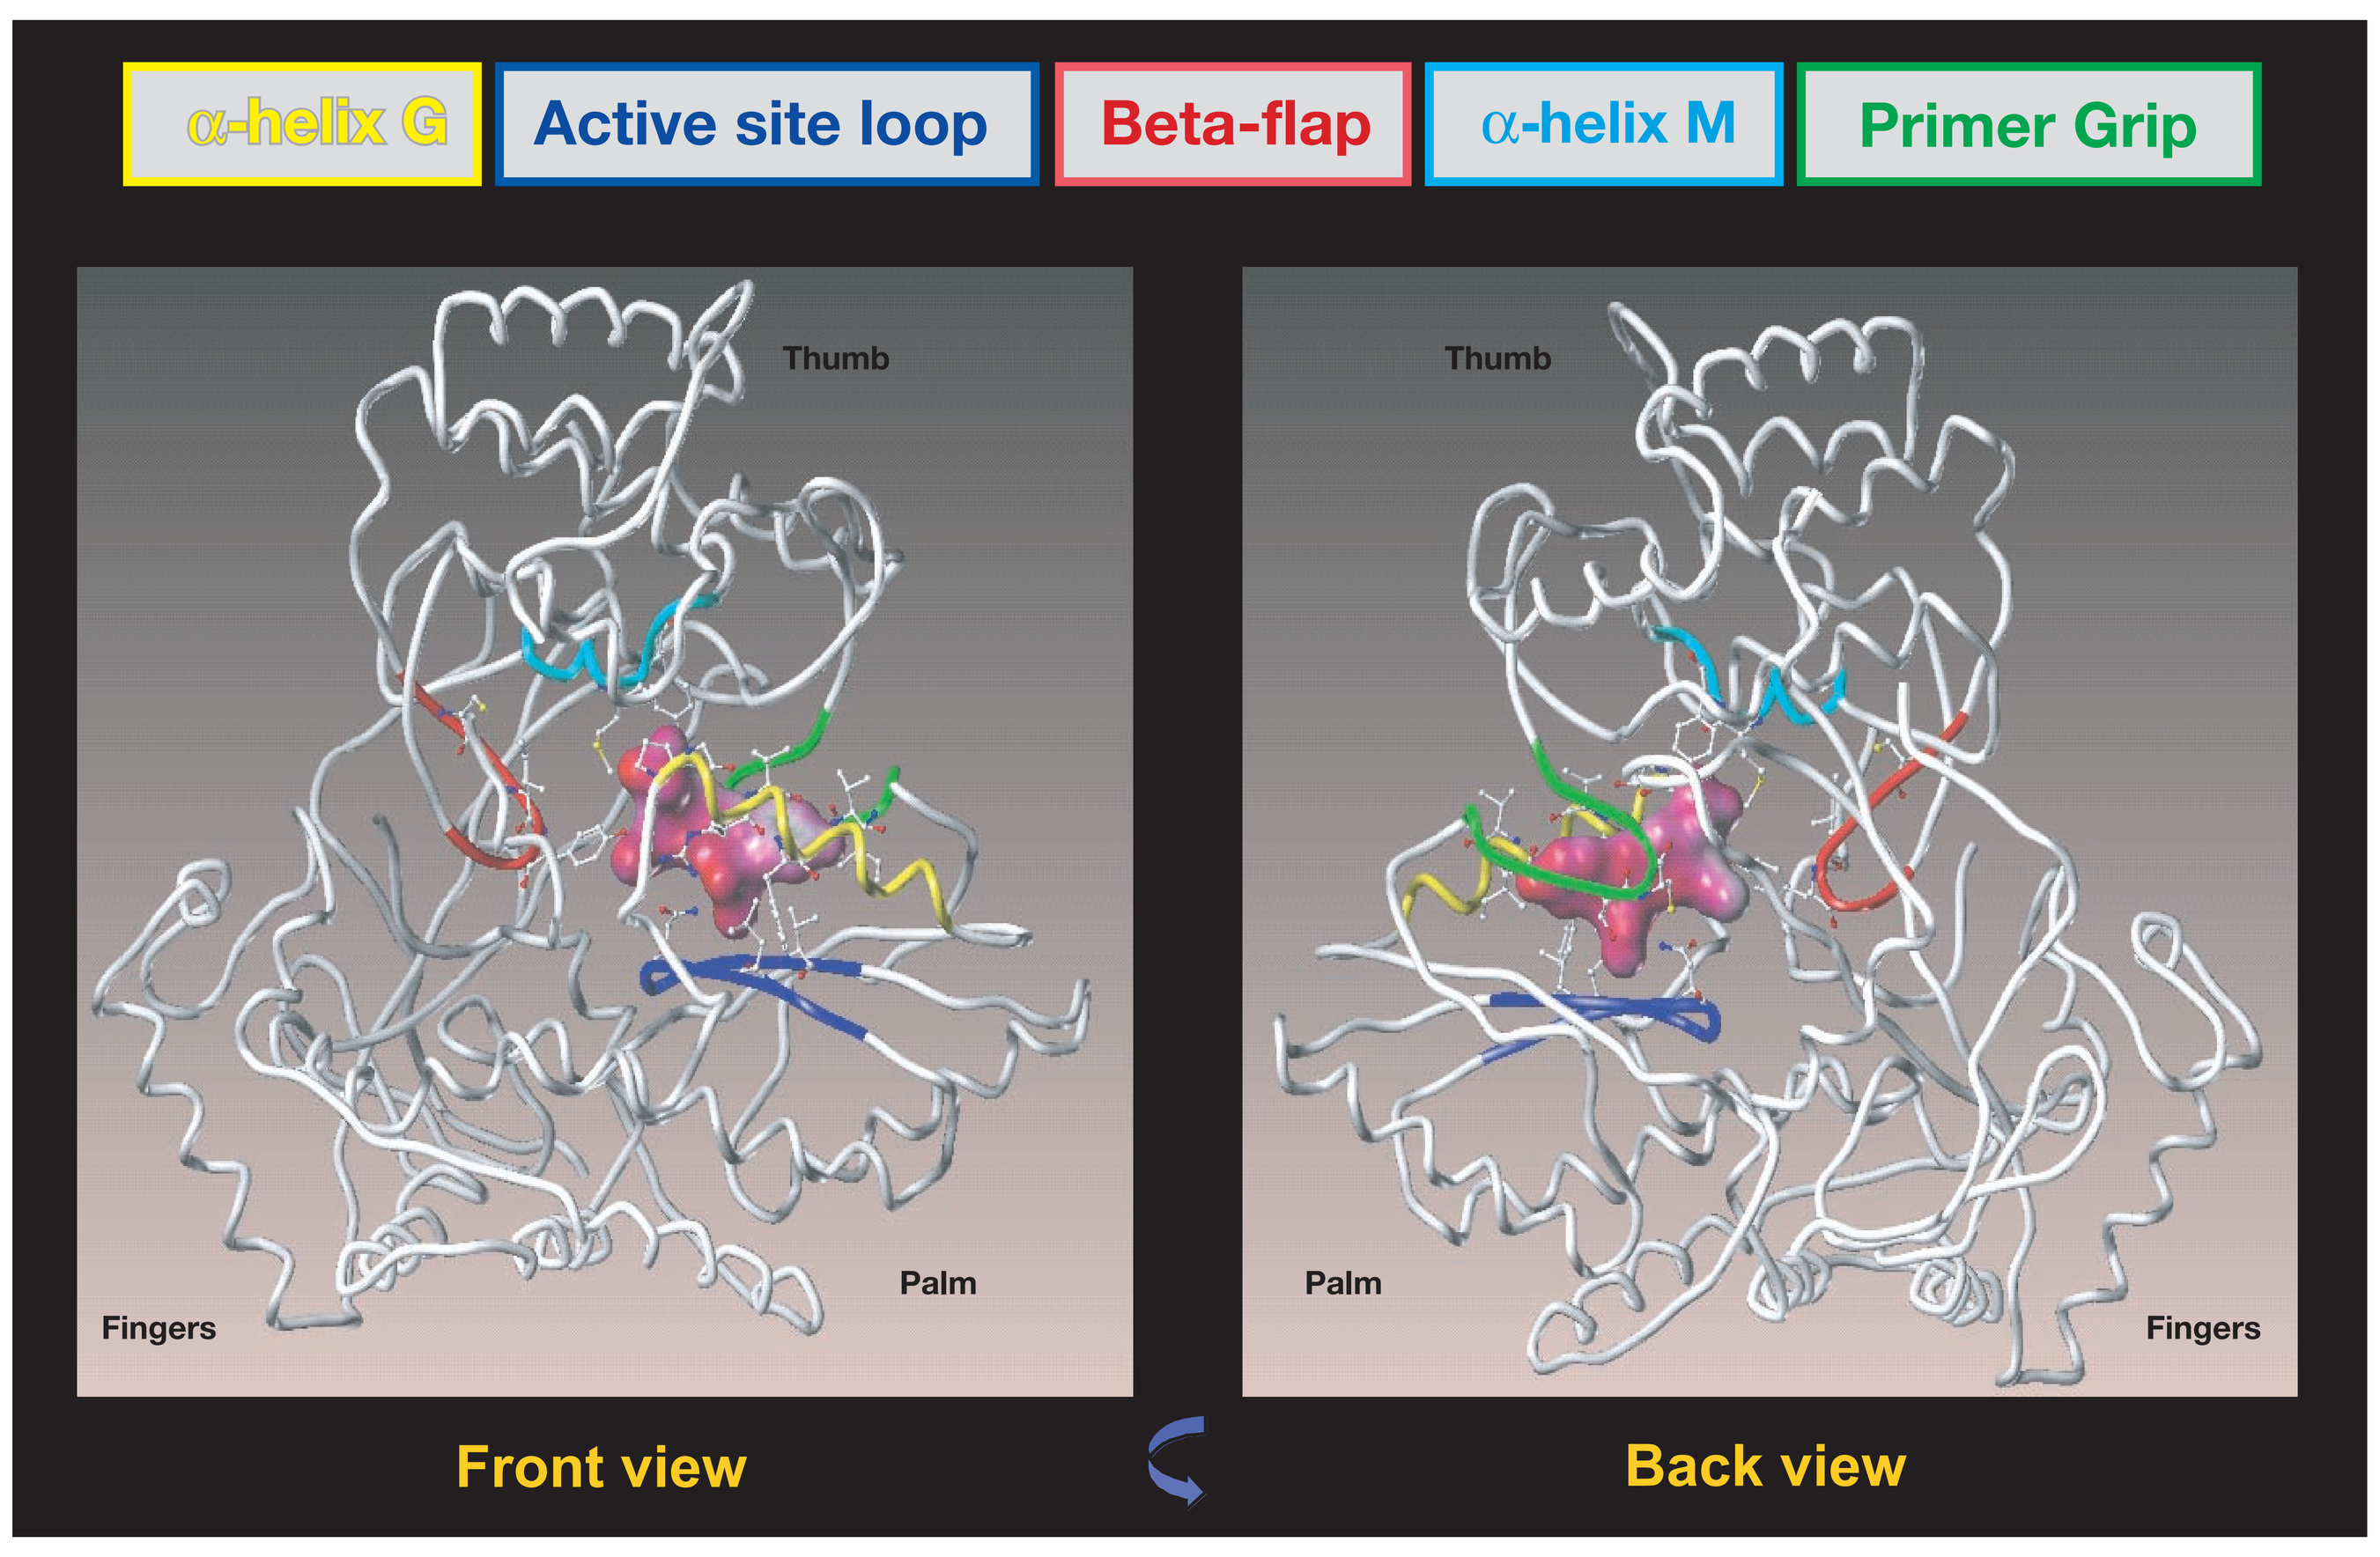

α-helix G
Active site loop
Beta-flap
α-helix M
Primer Grip
Thumb
Fingers
Palm
Thumb
Palm
Fingers
Front view
Back view

FIGURE 3b. Interactions between HCV-796 and Amino Acids

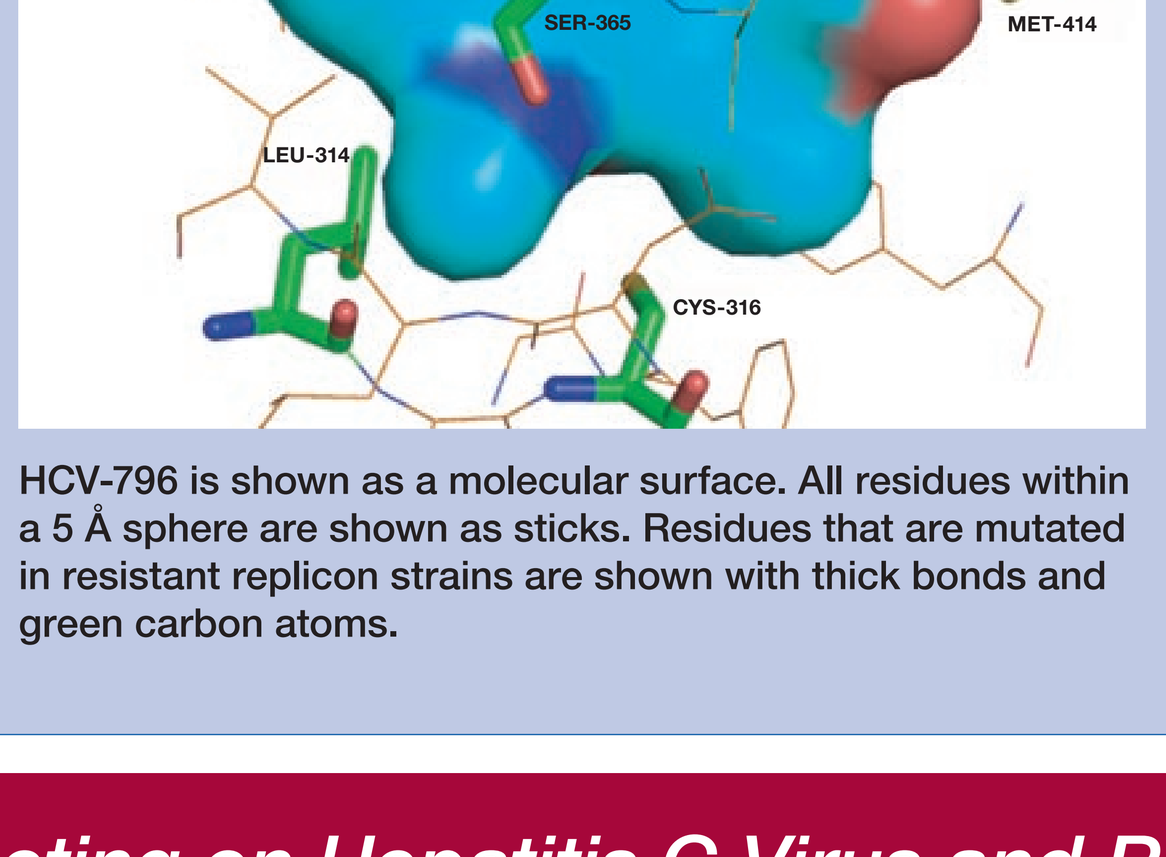

ILE-363
SER-365
MET-414
LEU-314
CYS-316

HCV-796 is shown as a molecular surface. All residues within a 5 Å sphere are shown as sticks. Residues that are mutated in resistant replicon strains are shown with thick bonds and green carbon atoms.

FIGURE 3c. Mutation C316F Clashes with HCV-796

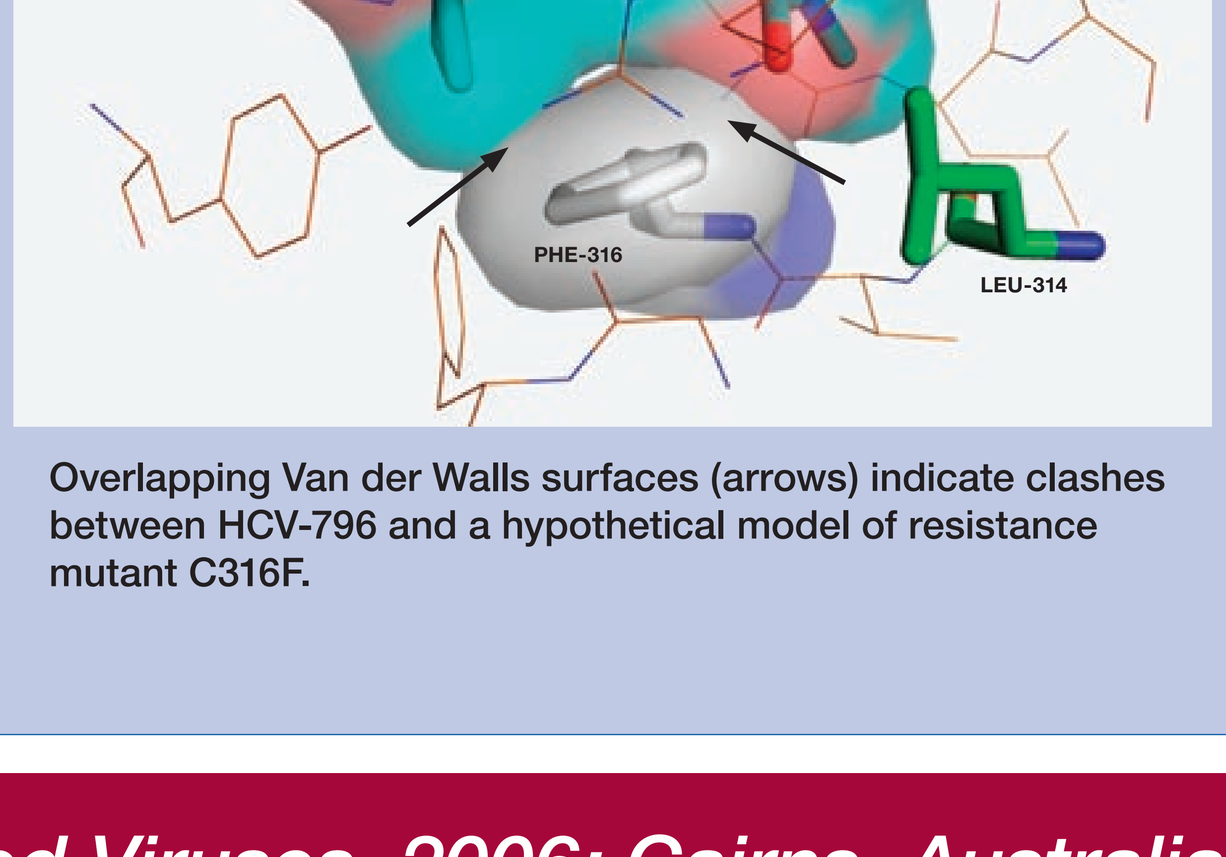

MET-414
SER-365
PHE-316
LEU-314

Overlapping Van der Walls surfaces (arrows) indicate clashes between HCV-796 and a hypothetical model of resistance mutant C316F.

Drug Susceptibility of Replicon Variants

Table 2a. Activity of HCV-796 Against HCV-796 Replicon Variants

Replicon Variant[a] HCV	HCV-796 RNA EC_{50} (nM) ± SD[b]	Fold Resistance	Viral Load Reduction[c]
1b, BB7	3.0 ± 1.0 (n=11)	–	1.9 ± 0.3
1b, BB7-L314F	4 ± 2 (n=4)	1	1.6 ± 0.3
1b, BB7-C316F	392 ± 209 (n=4)	130	0.8 ± 0.5
1b, BB7-C316Y	501 ± 291 (n=4)	166	0.9 ± 0.2
1b, BB7-C316N[d]	220 ± 110 (n=4)	26[d]	N/A
1b, BB7-C316S	30 ± 4 (n=4)	10	1.3 ± 0.1
1b, BB7-I363V	16 ± 5 (n=3)	5	1.4 ± 0.1
1b, BB7-S365A	124 ± 41 (n=4)	41	1.7 ± 0.1
1b, BB7-S365T	643 ± 168 (n=4)	212	0.6 ± 0.1
1b, BB7-S365L	N/A[e]	N/A[e]	N/A[e]
1b, BB7-S368F	5 ± 2 (n=4)	2	1.4 ± 0.3
1b, BB7-M414I	23 ± 3 (n=5)	8	1.5 ± 0.2
1b, BB7-M414T	3 ± 1 (n=4)	1	2.0 ± 0.2
1b, BB7-M414V	8 ± 1 (n=3)	3	1.5 ± 0.1

a: 1b, BB7 represents HCV genotype 1b, BB7 isolate. The nomenclature of the replicon NS5B variants (e.g. L314F) is expressed as the amino acid of the input replicon, amino acid position and amino acid substitution.
b: EC_{50} values were determined using the MDL LSW data analysis™. Inhibitory activity is expressed as mean EC_{50} ± standard deviation. n indicates number of determinations.
c: Viral load reduction was determined at 2240 nM HCV-796 in a 3-day assay. Data represent the mean log reduction of viral RNA ± standard deviation. Results represent at least 3 independent determinations.
d: The evaluation of 1b, BB7-C316N was evaluated in a separate laboratory. The EC_{50} for HCV-796 in 1b, BB7 was 8.6 ± 4 (n=14), which was used to calculate the fold resistance for 1, BB7-C316N.
e: Replicon variant S365L failed to establish a stable cell line upon selection with G418.

Colony Formation Efficiency and Growth Kinetics of Replicon Variants

Table 3a. Colony Formation Efficiency of Replicon Variants in Huh-7 cells

Replicon Variant	CFU/µg RNA
1b, BB7 control	20,000
L314F	1500
C316F	3,000 - 5,000
C316S	3,000 - 5,000
I363V	100
S365A	1000
S365T	120
S365L	20[a]
M414V	500
M414T	3000

a: did not survive G418 selection

Table 3b. Steady State Viral Load in HCV-796 Associated Replicon Variants

Replicon Variant	Steady-state Viral Load (HCV copies/µg) ± SD
1b, BB7	$1.8 \pm 1.1 \times 10^8$
796R (0.1 µM)	$2.9 \pm 0.5 \times 10^8$
796R (1 µM)	$2.4 \pm 2.0 \times 10^8$
796R (10 µM)	$4.1 \pm 1.7 \times 10^8$
1b, BB7-L314F	$0.3 \pm 0.1 \times 10^8$
1b, BB7-C316F	$1.0 \pm 0.2 \times 10^8$
1b, BB7-C316Y	$1.3 \pm 0.6 \times 10^8$
1b, BB7-C316N	N/A
1b, BB7-C316S	$1.3 \pm 0.7 \times 10^8$
1b, BB7-I363V	$0.2 \pm 0.1 \times 10^8$
1b, BB7-S365A	$1.2 \pm 0.3 \times 10^8$
1b, BB7-S365T	$1.3 \pm 0.6 \times 10^8$
1b, BB7-S365L	N/A[a]
1b, BB7-S368F	$2.6 \pm 1.2 \times 10^8$
1b, BB7-M414I	$1.3 \pm 0.5 \times 10^8$
1b, BB7-M414T	$1.5 \pm 0.7 \times 10^8$
1b, BB7-M414V	$0.4 \pm 0.1 \times 10^8$

a: Replicon variant S365L failed to establish a stable cell line upon selection with G418; NA = not available.

Drug Susceptibility of HCV-796 Replicon Variants with Other Antiviral Agents

Table 4. Activities of Antiviral Agents Against HCV-796 Associated Replicon Variants

Compound	Replicon	EC_{50} (µM or pg/ml) ± SD	Fold Resistance
PegIFN α-2b[a]	1b, BB7 (WT)	7.0 ± 0.5 (n=3)	-
	C316Y	8.0 ± 3.9 (n=3)	1.1
RBV	1b, BB7 (WT)	132.6 ± 40.5 (n=5)	-
	C316Y	200.9 ± 36.7 (n=3)	1.8
HCV-371[b]	1b, BB7 (WT)	12.2 ± 1.8 (n=2)	-
	C316Y	10.8 ± 0.8 (n=2)	0.9

a: expressed in pg/ml
b: a pyranoindole HCV NS5B inhibitor that binds to a different site from HCV-796

CONCLUSIONS

- Replicon cells with reduced susceptibility to HCV-796 were generated through prolonged treatment with HCV-796 followed by G418 enrichment.
- The selected pools of replicon cells exhibited 23- to >6800-fold reduced susceptibility to HCV-796.
- Sequencing of NS5B in the replicon variants mapped amino acid changes within the drug-binding pocket, validating the enzyme target of HCV-796.
- The impact of amino acid changes on resistance was examined in replicon variants bearing single amino acid mutations. C316F/Y and S365T/A are identified as the key amino acid mutations responsible for the resistance *in vitro*.
- Most of the replicon variants have lower colony formation efficiency. Some variants also have lower steady-state levels of HCV RNA.
- Replicon variants that have reduced susceptibily remain sensitive to the inhibition by pegylated interferon, ribavirin, and another non-nucleoside HCV polymerase inhibitor.

Wyeth ViroPharma Incorporated

13th International Meeting on Hepatitis C Virus and Related Viruses, 2006; Cairns, Australia, 27-31 August, 2006